NKTR-118 Significantly Reverses Opioid-Induced Constipation

NEKTAR

Lynn Webster, MD [1] and Sunita Dhar, MD[2] on behalf of the NKTR-118 Study 07-IN-NX003 Cooperative Group,
Lifetree Research[1], Salt Lake City, UT; Nektar Therapeutics[2], San Carlos, CA

Abstract

NKTR-118, a novel PEGylated naloxol conjugate, is an oral, once daily investigational agent in development for treatment of opioid-induced constipation (OIC). NKTR-118 blocks peripheral mu opioid receptors in the gastrointestinal tract to reverse OIC, but was designed to avoid entry into the brain to maintain centrally mediated analgesia. The objective of this phase 2 study was to determine the safety and efficacy of NKTR-118 in patients with opioid-induced constipation. Eligible patients had OIC (fewer than three spontaneous bowel movements [SBMs] per week) and were on a stable opioid regimen of 30 to 1000 morphine equivalent units per day for a minimum of two weeks. A total of 208 patients were randomized 1:1 into 3 sequential cohorts receiving a once daily oral dose of placebo for one week (single-blind) followed by a once daily oral dose of placebo or NKTR-118 at doses of 5, 25 or 50 mg for four consecutive weeks. The primary endpoint was change from baseline in SBMs per week during the first week of double-blind study treatment.

Patients receiving 25 mg NKTR-118 had a mean of 5.0 SBMs during the first week of treatment as compared to 1.4 SBMs per week at baseline versus 3.1 and 1.2 SBMs per week during the same periods for the placebo group (p=0.002). Patients receiving 50 mg NKTR-118 had a mean of 6.0 SBMs during the first week of treatment as compared to 1.6 SBMs per week during baseline versus 3.3 and 1.3 SBMs during the same periods for the placebo group (p=0.0001). The increase in SBMs versus placebo was maintained over the 28-day treatment period for both 25 mg and 50 mg dose groups (p=0.002 and p<0.0001 respectively). Results from the 5 mg dose group compared to placebo were not statistically significant. There was no evidence of opioid withdrawal, reversal of analgesia or increase in opioid use at any dose tested. The only study drug related SAE was abdominal cramping that led to hospitalization in one subject (50 mg). The most frequent side effects were abdominal cramping, diarrhea, nausea and vomiting, occurring more often in the 50 mg cohort.

Results of this study showed that oral NKTR-118 restores gastrointestinal function by increasing the frequency of SBMs to essentially normal in patients with OIC. These results were highly statistically significant for both the 25 mg and 50 mg once daily doses. There was no CNS withdrawal or reversal of analgesia.

Nektar Platform

Nektar Therapeutics proprietary polymer technology has been successfully used to improve the pharmacokinetics and pharmacodynamics of several marketed therapeutic agents (eg, Neulasta®, Cimzia®, PEG-INTRON™, and PEGASYS®). Nektar Therapeutics is developing NKTR-118, a PEGylated conjugate of naloxone, for the treatment of opioid-induced constipation.

Background

Constipation is commonly associated with use of opiates for chronic pain and can significantly reduce quality of life. Dietary changes, exercise and stimulant laxatives do not provide effective relief. NKTR-118, a PEGylated conjugate of naloxol is under development for the treatment of opioid-induced constipation. Naloxone is the gold standard opioid antagonist used to reverse the effects of opioids. However, when used as a treatment for opioid induced constipation, it can also enter the brain and may lead to reversal of analgesia or opiate withdrawal. NKTR-118 has been specifically engineered to act peripherally to normalize bowel function while maintaining opioid induced central analgesia.

Methods

Eligible patients had OIC (<3 SBMs per week) and were on a stable opioid regimen of 30 to 1000 MEU per day for a minimum of 2 weeks. A total of 208 patients were randomized 1:1 into 3 sequential cohorts receiving a once daily oral dose of placebo for one week (single-blind) followed by a once daily oral dose of placebo or NKTR-118 at doses of 5, 25 or 50 mg for four consecutive weeks.

Figure 1. Study Schema:

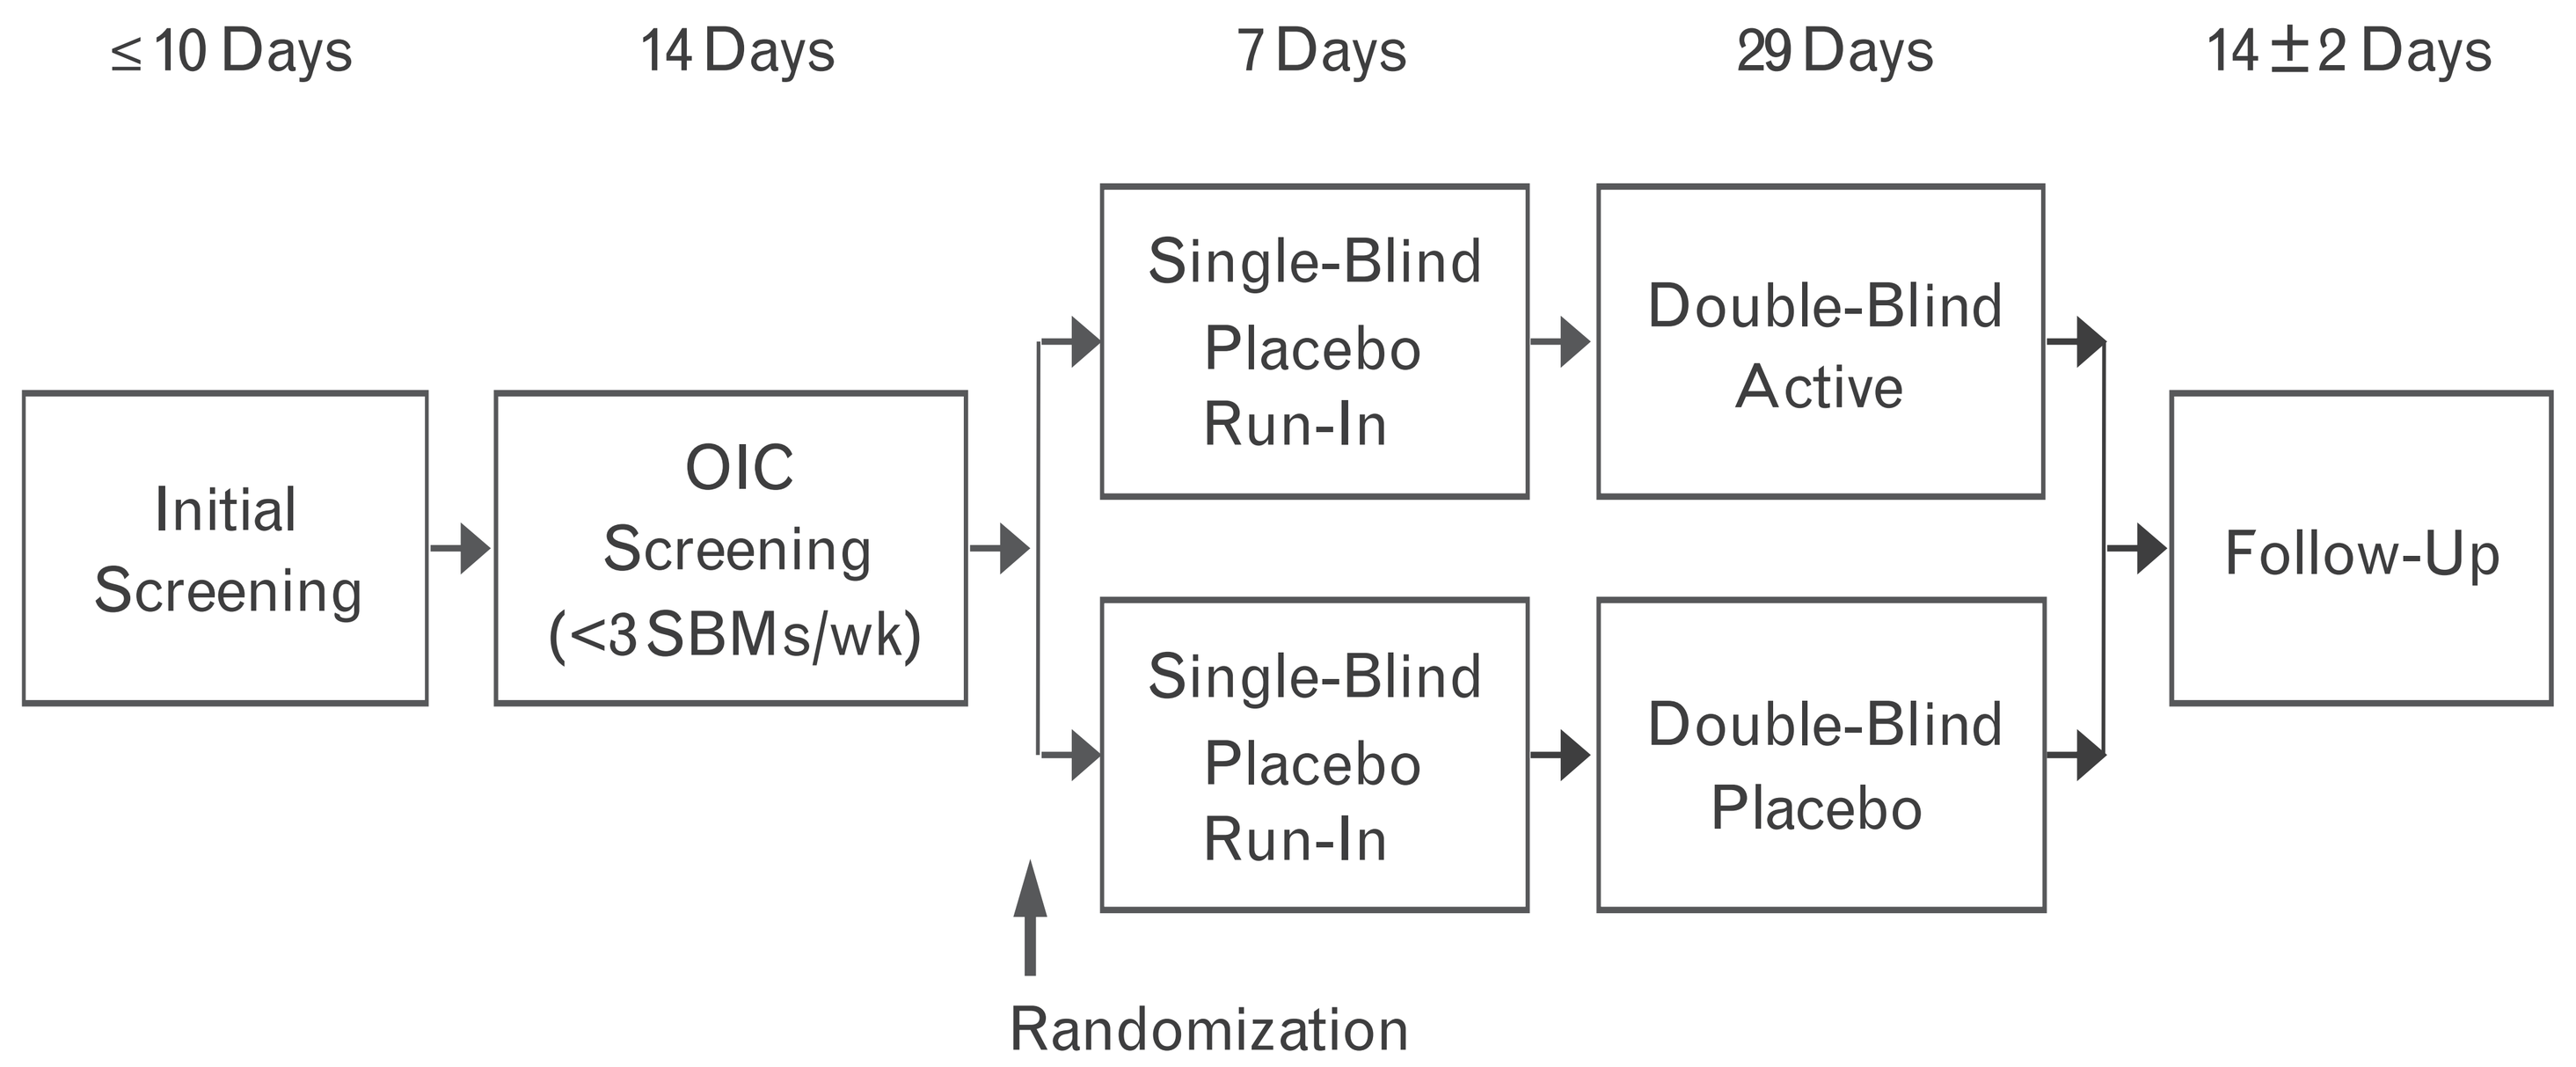
≤ 10 Days
14 Days
7 Days
29 Days
14 ± 2 Days
Initial Screening
OIC Screening (<3 SBMs/wk)
Single-Blind Placebo Run-In
Double-Blind Active
Single-Blind Placebo Run-In
Double-Blind Placebo
Follow-Up
Randomization

Results

Superior Activity of NKTR-118 versus Placebo:
Significant change from baseline in SBMs/week during the first week of double-blind (DB) study treatment for both the 25 mg and 50 mg dose groups as compared to placebo

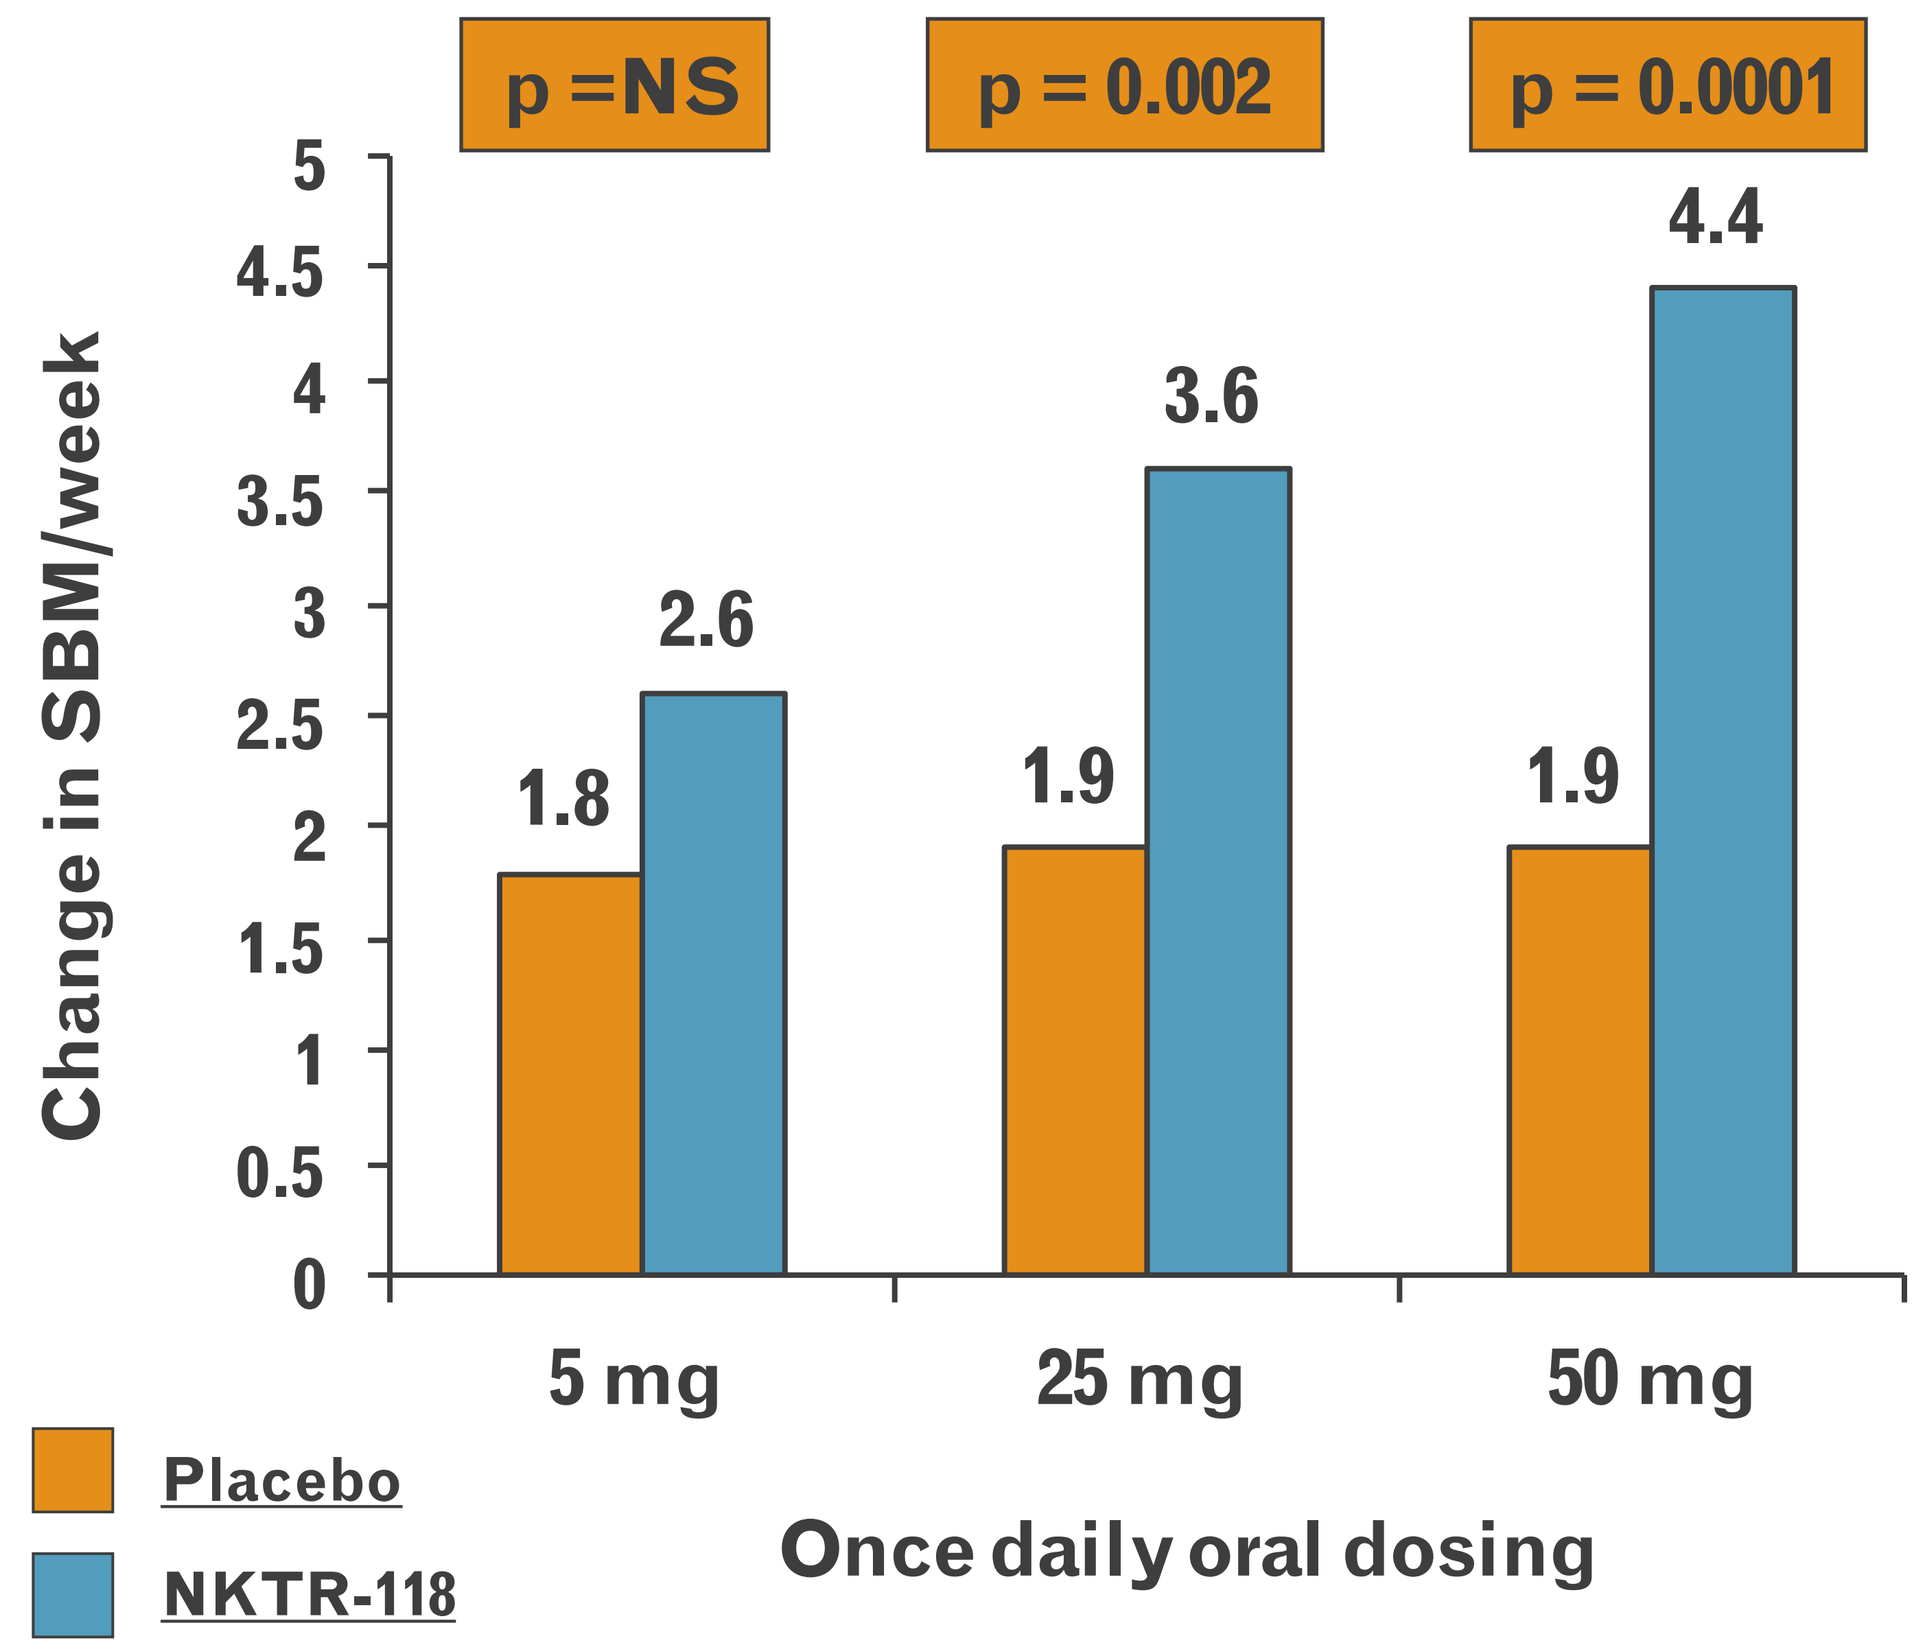
p =NS
p = 0.002
p = 0.0001
Change in SBM/week
5
4.5
4
3.5
3
2.5
2
1.5
1
0.5
0
1.8
2.6
1.9
3.6
1.9
4.4
5 mg
25 mg
50 mg
Placebo
NKTR-118
Once daily oral dosing

Significantly higher proportion of responders averaged over the 4 wk DB treatment period for NKTR-118 patients in both the 25 mg group and the 50 mg group

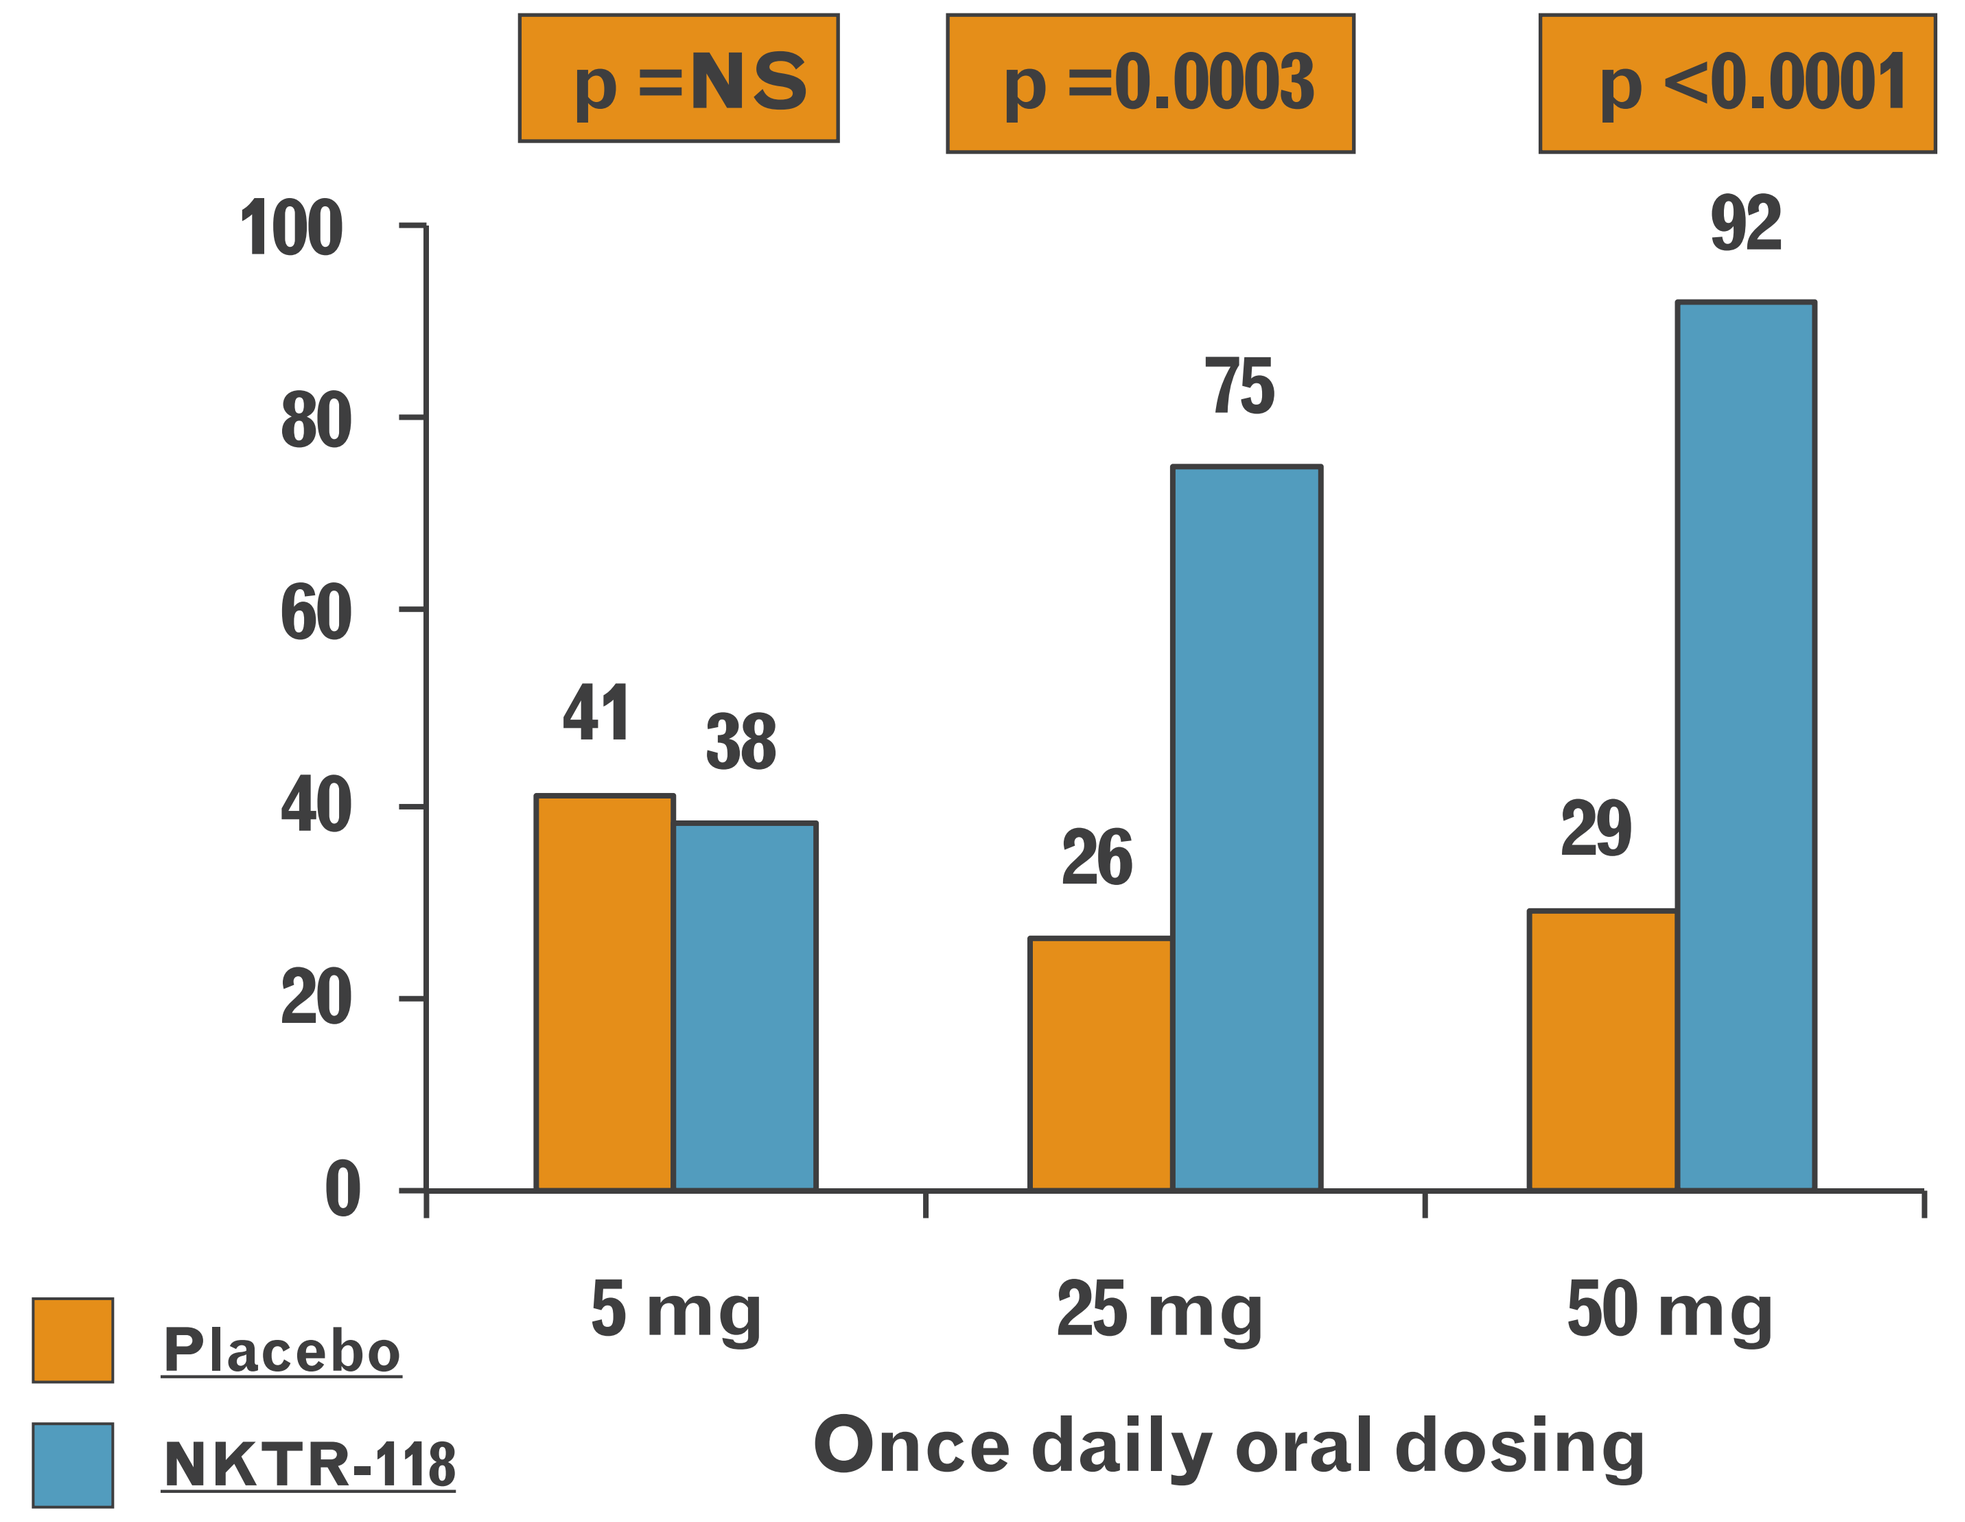
p =NS
p =0.0003
p <0.0001
100
80
60
40
20
0
41
38
26
75
29
92
5 mg
25 mg
50 mg
Placebo
NKTR-118
Once daily oral dosing

No Reversal of Analgesia:
Daily Opioid Use Unchanged
(50 mg group shown – similar results for 5 and 25 mg dose groups)

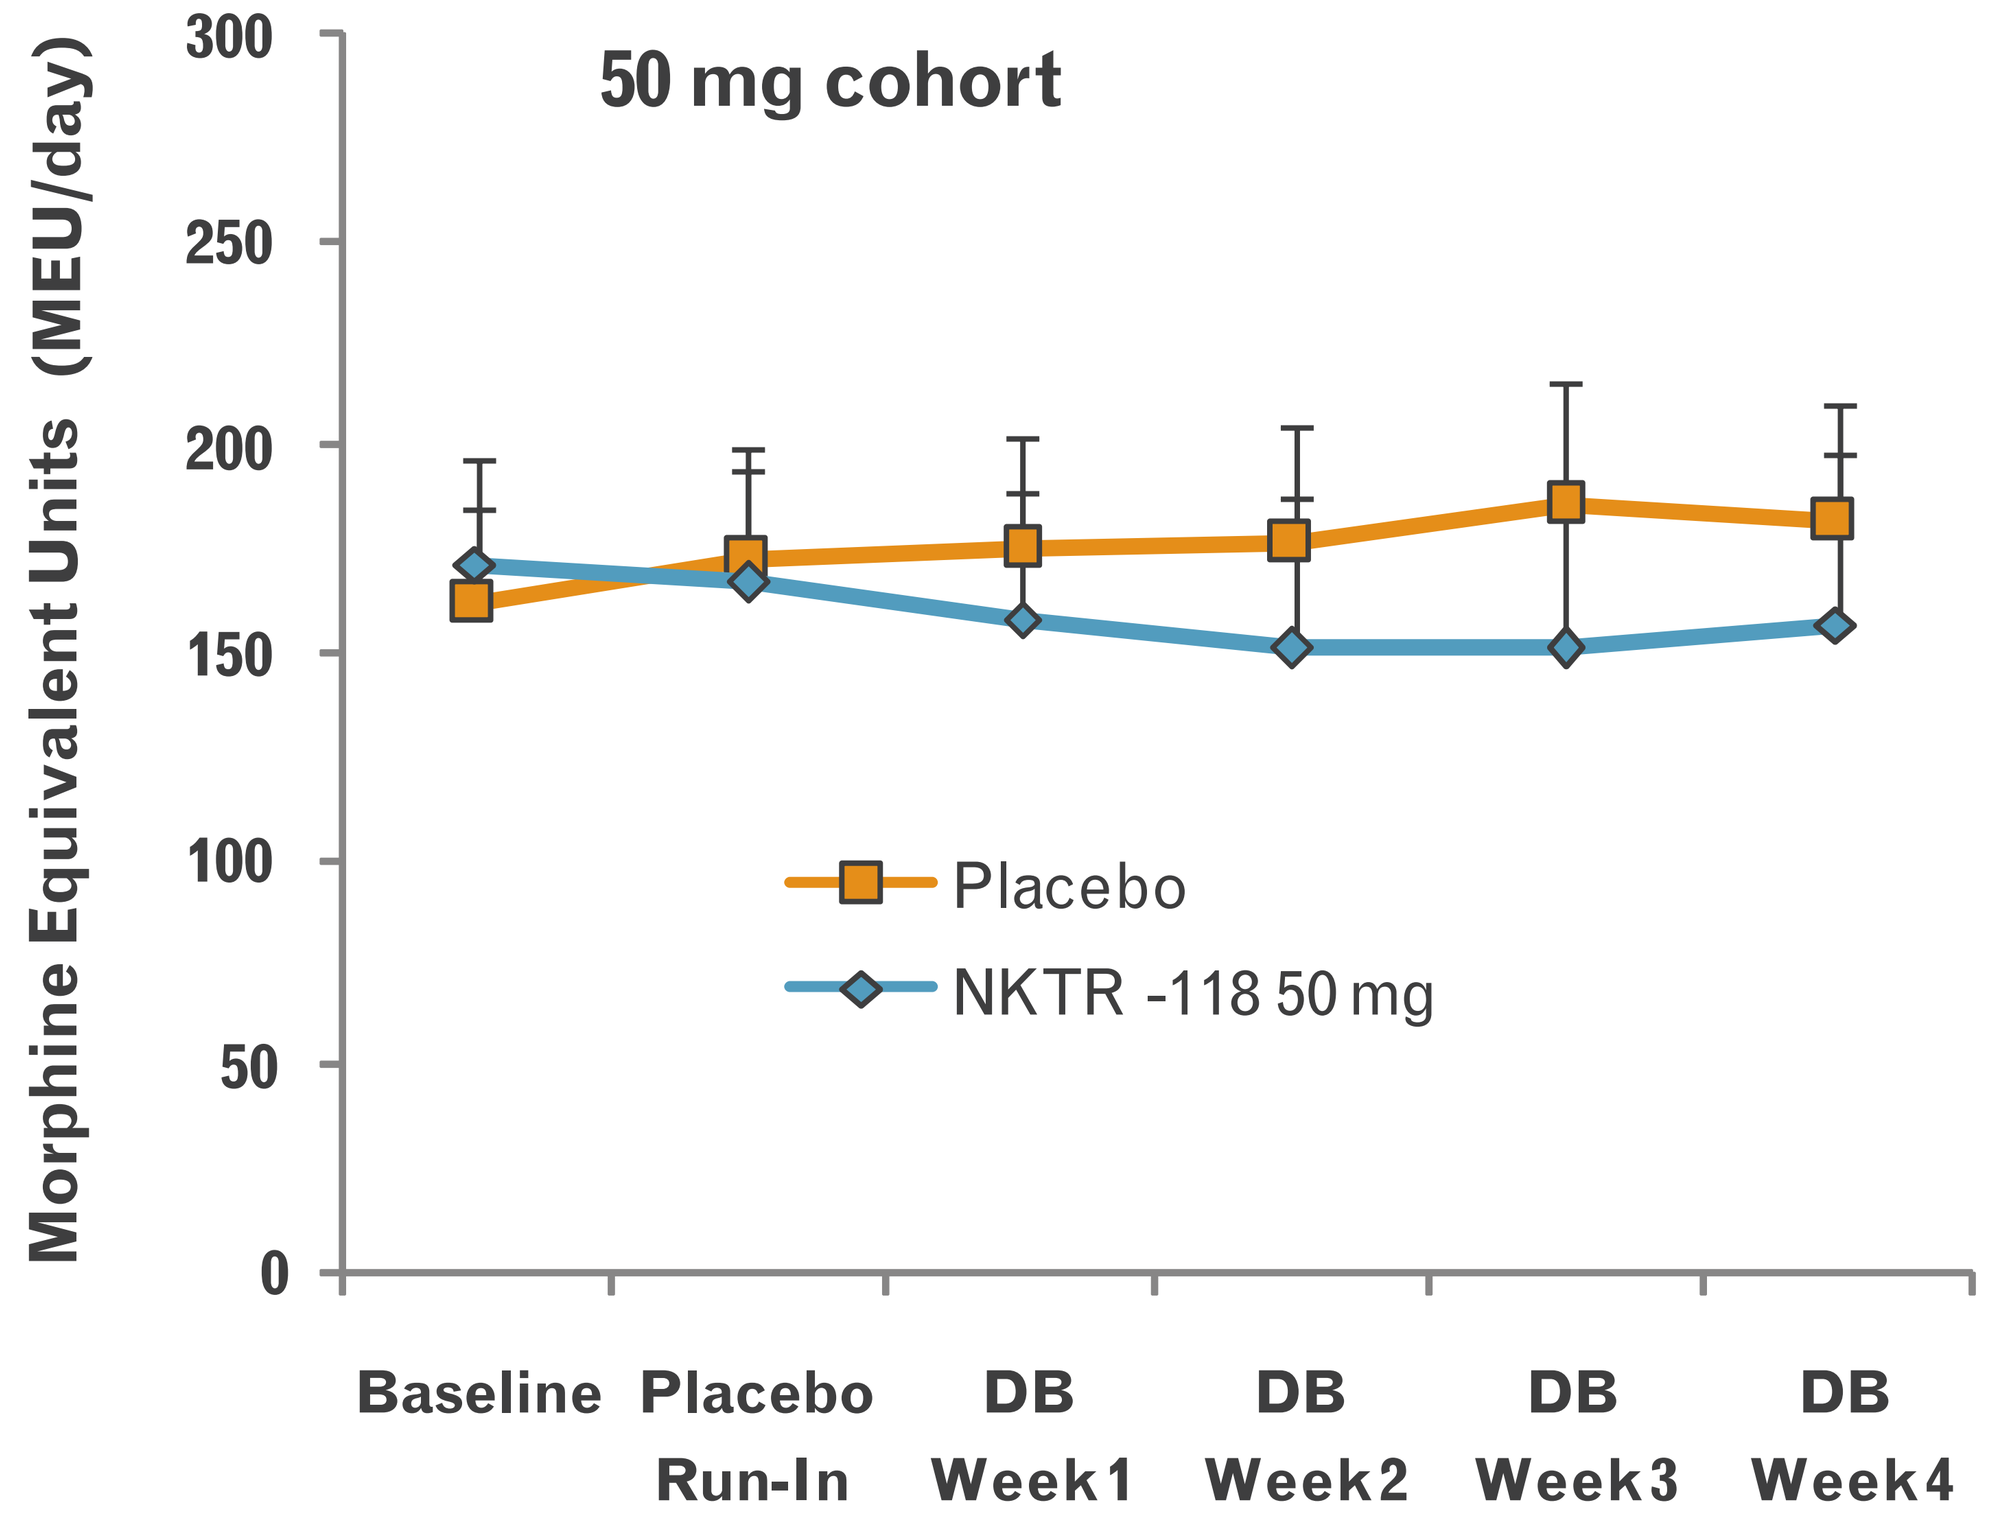
Morphine Equivalent Units (MEU/day)
300
250
200
150
100
50
0
50 mg cohort
Placebo
NKTR -118 50 mg
Baseline
Placebo Run-In
DB Week1
DB Week2
DB Week3
DB Week4

Conclusion

- Oral NKTR-118 restores gastrointestinal function by increasing the frequency of SBMs to essentially normal in patients with OIC.
- There was no reversal or reduction of opioid-mediated analgesia at the 5 mg, 25 mg, or 50 mg dose groups (no shift in pain scores or increase in opiate use).
- NKTR-118 was well tolerated with the most commonly reported side effects being dose dependent GI –related effects.

References:

Pappagallo M. Incidence, prevalence and management of opioid bowel dysfunction. Am J Surg 2001;182(5A Suppl):11s-18s.
Reisine T, Pasternak G. Opioid analgesics and antagonists In Hardman JG, Limbird LE, Molinoff PB, Ruddon RW, Gilman AG, eds. Goodman & Gilman's The pharmacological basis of therapeutics. 9th ed. New York, NY: McGraw-Hill;1996:521-555.
Neumann TA, Van Paaschen H, Marcantonio A, Song D, Morrison PH, Eldon MA, Jugler AR. 2007 Clinical Investigation of NKTR-118 as a selective oral peripheral opioid antagonist, Abstr. 27. In Programs and Abstracts of American Academy of Pain Management (AAPM), Las Vegas NV, USA.

This study was supported by Nektar Therapeutics, San Carlos, CA

© 2009 Nektar Therapeutics.